L957

Improved Metabolic Profile with Replacement of Stavudine by Tenofovir DF After 6 Years of a LPV/r-Based Regimen

B da Silva[1], M Albrecht[2], C Benson[3], J Eron[4], R Gulick[5], M Glesby[5], C Hicks[6], H Kessler[7], R Murphy[8], M Thompson[9], AC White[10], P Wolfe[11], F McMillan[1], K Niemi[1], M King[1], G Hanna[1]. [1]Abbott, Abbott Park, IL USA; [2]Harvard U, Boston, MA USA; [3]UCSD, San Diego, CA, USA; [4]UNC, Chapel Hill, NC, USA; [5]Cornell U, NY, NY, USA; [6]Duke U, Durham, NC, USA; [7]Rush Med College, Chicago, IL, USA; [8]Northwestern U, Chicago, IL, USA; [9]ARCA, Atlanta, GA, USA; [10]Baylor College of Med., Houston, TX, USA; [11]Pacific Oaks Research, Beverly Hills, CA, USA

Background

Stavudine has been associated with the development of dyslipidemia, specifically hypercholesterolemia and hypertriglyceridemia [1], and replacement of stavudine with tenofovir disoproxil fumarate (tenofovir DF) decreased lipid levels [2,3]. Similar findings are noted when stavudine is substituted by abacavir [4].

Apolipoprotein CIII is an inhibitor of lipoprotein lipase and as such, inhibits the breakdown of triglyceride-rich lipoproteins. Increased levels of apolipoprotein CIII is characteristic of patients with hypertriglyceridemia. Apolipoprotein CIII levels decrease with both statin and fibrate therapy in patients with hypertriglyceridemia. [5,6]

Adiponectin is an adipokine that plays a role in insulin sensitivity, with decreased adiponectin in insulin resistance and type 2 diabetes. [7] In mouse models, infusions of adiponectin and leptin resulted in improvement of hyperglycemia and hyperinsulinemia. [8] Finally, adiponectin levels are low in patients receiving stavudine, but appear to be increased with lopinavir/ritonavir (LPV/r) administration in HIV-negative subjects. [9,10]

Leptin is an adipocyte-derived hormone that acts as a major regulator for food intake and energy homeostasis. [11] Deficiency of leptin results in obesity and type 2 diabetes. Leptin levels in HIV-infected subjects are lower in those with lipoatrophy, compared to those without body fat changes. [12]

Highly sensitive C-reactive protein (hsCRP) levels are predictive of cardiovascular disease risk, while homocysteine levels correlate with vascular disease in HIV-1-uninfected individuals. In addition to information provided by LDL levels, hsCRP levels may be used to predict cardiovascular risk with levels <1 mg/dL, correlating with low cardiovascular risk. [13,14]

In this study, we examine whether replacing stavudine with tenofovir DF can result in metabolic benefits assessed by measurement of total cholesterol, triglycerides, apolipoprotein CIII, adiponectin, leptin, homocysteine and hsCRP for subjects who have received LPV/r + stavudine + lamivudine for over 6 years.

Methods

- In Study 720, antiretroviral-naive HIV-1-infected subjects were randomized to one of three blinded doses of LPV/r (200/100 mg BID, 400/100 mg BID or 400/200 mg BID), together with stavudine (40 mg BID) and lamivudine (150 mg BID). After 48 weeks, all subjects converted to open-label LPV/r 400/100 mg BID.
- Subjects treated with LPV/r, stavudine, and lamivudine for over 6 years were given the option of replacing stavudine with tenofovir DF. Subjects who had previously replaced stavudine with zidovudine were allowed to switch from zidovudine to tenofovir DF.
- Fasting laboratory measurements obtained prior to the switch to tenofovir DF and at 12 and 24 weeks after the switch included total cholesterol (TC), HDL cholesterol, LDL cholesterol, triglycerides (TG), glucose, apolipoprotein AI (Apo-AI), apolipoprotein CIII (Apo-CIII), adiponectin, leptin, highly sensitive C-reactive protein (hsCRP), and homocysteine. HOMA-IR index (insulin [mciu/mL] X glucose [mmol/L] / 22.5) was also assessed.
- Changes in laboratory measurements were assessed using a paired *t* test.
- Plasma HIV-1 RNA levels and CD4 cell counts before and after the switch to tenofovir DF were also assessed.

Results

Subject Disposition

- 37 subjects treated with LPV/r + stavudine + lamivudine for a median of 6.6 years substituted stavudine with tenofovir DF and were treated for at least 12 weeks. 23 of these subjects were treated for at least 24 weeks with tenofovir DF. Three subjects who replaced zidovudine with tenofovir DF were not included in the analysis.
- Plasma HIV-1 RNA levels and CD4 cell counts remained stable after the switch to tenofovir DF: 35/37 (95%) subjects had HIV-1 RNA <50 copies/mL both prior to and 12 weeks after the switch. Mean CD4 count was 768 cells/mm^3 prior to switch and 824 cells/mm^3 12 weeks after the switch (p=0.13).

Changes in Lipid Levels

- Significant decreases in TC and TG were observed after 12 weeks of tenofovir DF treatment, with continued decreases through 24 weeks (Figure 1). Mean TC values decreased from 220 mg/dL (5.7 mmol/L) prior to the switch to 204 mg/dL (5.3 mmol/L) after 12 weeks of tenofovir DF (p=0.009). Mean TG values decreased from 373 mg/dL (4.2 mmol/L) prior to the switch to 254 mg/dL (2.9 mmol/L) after 12 weeks of tenofovir DF (p=0.023).
- Mean HDL cholesterol and LDL cholesterol values prior to the switch to tenofovir DF were 43 mg/dL (1.1 mmol/L) and 124 mg/dL (3.2 mmol/L), respectively. No changes in HDL cholesterol and LDL cholesterol values were observed after the switch to tenofovir DF (Figure 1).

Figure 1. Changes in Lipid Levels After Switch to Tenofovir DF

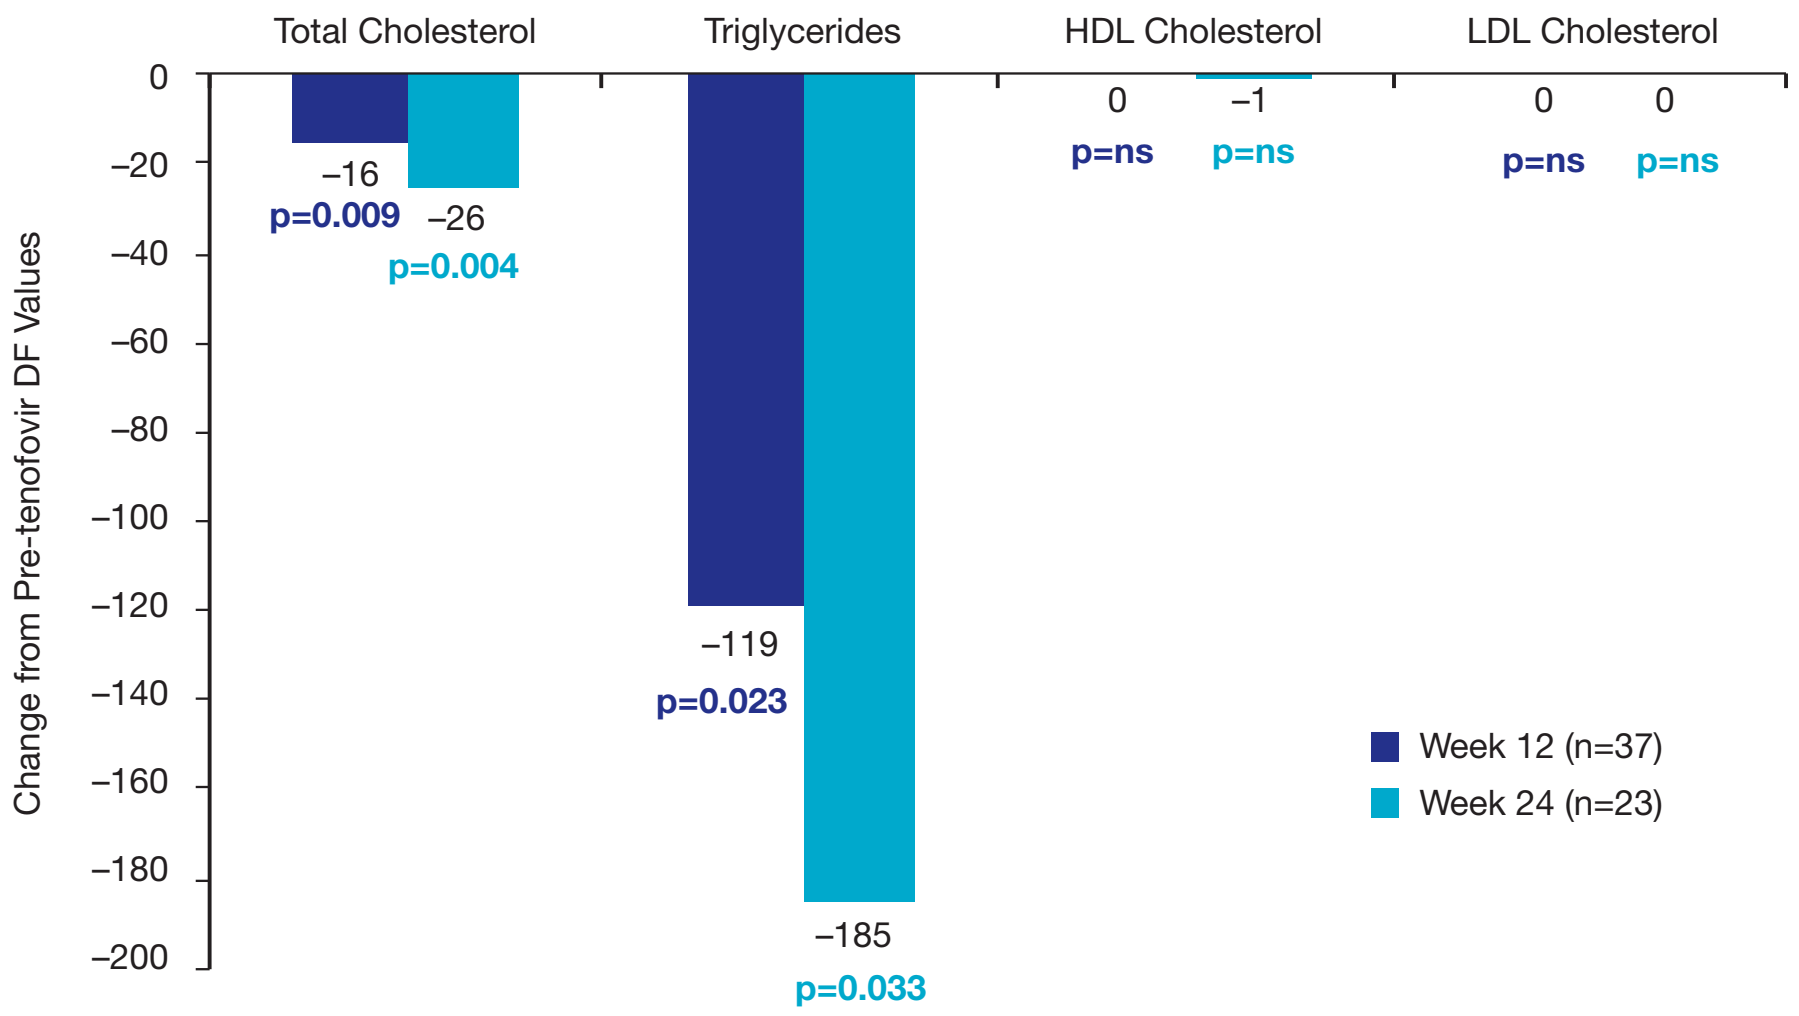

Total Cholesterol
Triglycerides
HDL Cholesterol
LDL Cholesterol
Change from Pre-tenofovir DF Values
0
–20
–40
–60
–80
–100
–120
–140
–160
–180
–200
–16
p=0.009
–26
p=0.004
–119
p=0.023
–185
p=0.033
0
p=ns
–1
p=ns
0
p=ns
0
p=ns
Week 12 (n=37)
Week 24 (n=23)

p-values represent comparison with values prior to switch to tenofovir DF

- 12 subjects were using lipid-lowering agents at the time of the switch to tenofovir DF. When these 12 subjects were excluded from the analysis, results were generally similar to the overall results. Median decrease after 12 and 24 weeks of tenofovir DF for TC were 18 mg/dL (0.47 mmol/L) and 25 mg/dL (0.65 mmol/L), respectively, and for TG were 85 mg/dL (0.96 mmol/L) and 162 mg/dL (1.83 mmol/L), respectively, with no significant changes in HDL and LDL cholesterol.

Changes in Other Metabolic Parameters

- Mean Apo-CIII values prior to the switch to tenofovir DF were 19.2 mg/dL. Significant decreases in mean Apo-CIII levels were observed 12 and 24 weeks following the switch to tenofovir DF (Figure 2).
- Mean adiponectin values prior to the switch to tenofovir DF were 3.8 mcg/dL. Significant increases in mean adiponectin levels were observed 12 weeks after the switch to tenofovir DF (Figure 2).
- Mean leptin values prior to the switch to tenofovir DF were 5.3 ng/mL. Significant increases in mean leptin levels were observed 24 weeks after the switch to tenofovir DF (Figure 2).
- Mean insulin values prior to the switch to tenofovir DF were 11.7 mciu/mL. Significant decreases in mean insulin levels were observed 24 weeks after the switch to tenofovir DF (Figure 2).

Figure 2. Significant Changes in Metabolic Parameters After Switch to Tenofovir DF

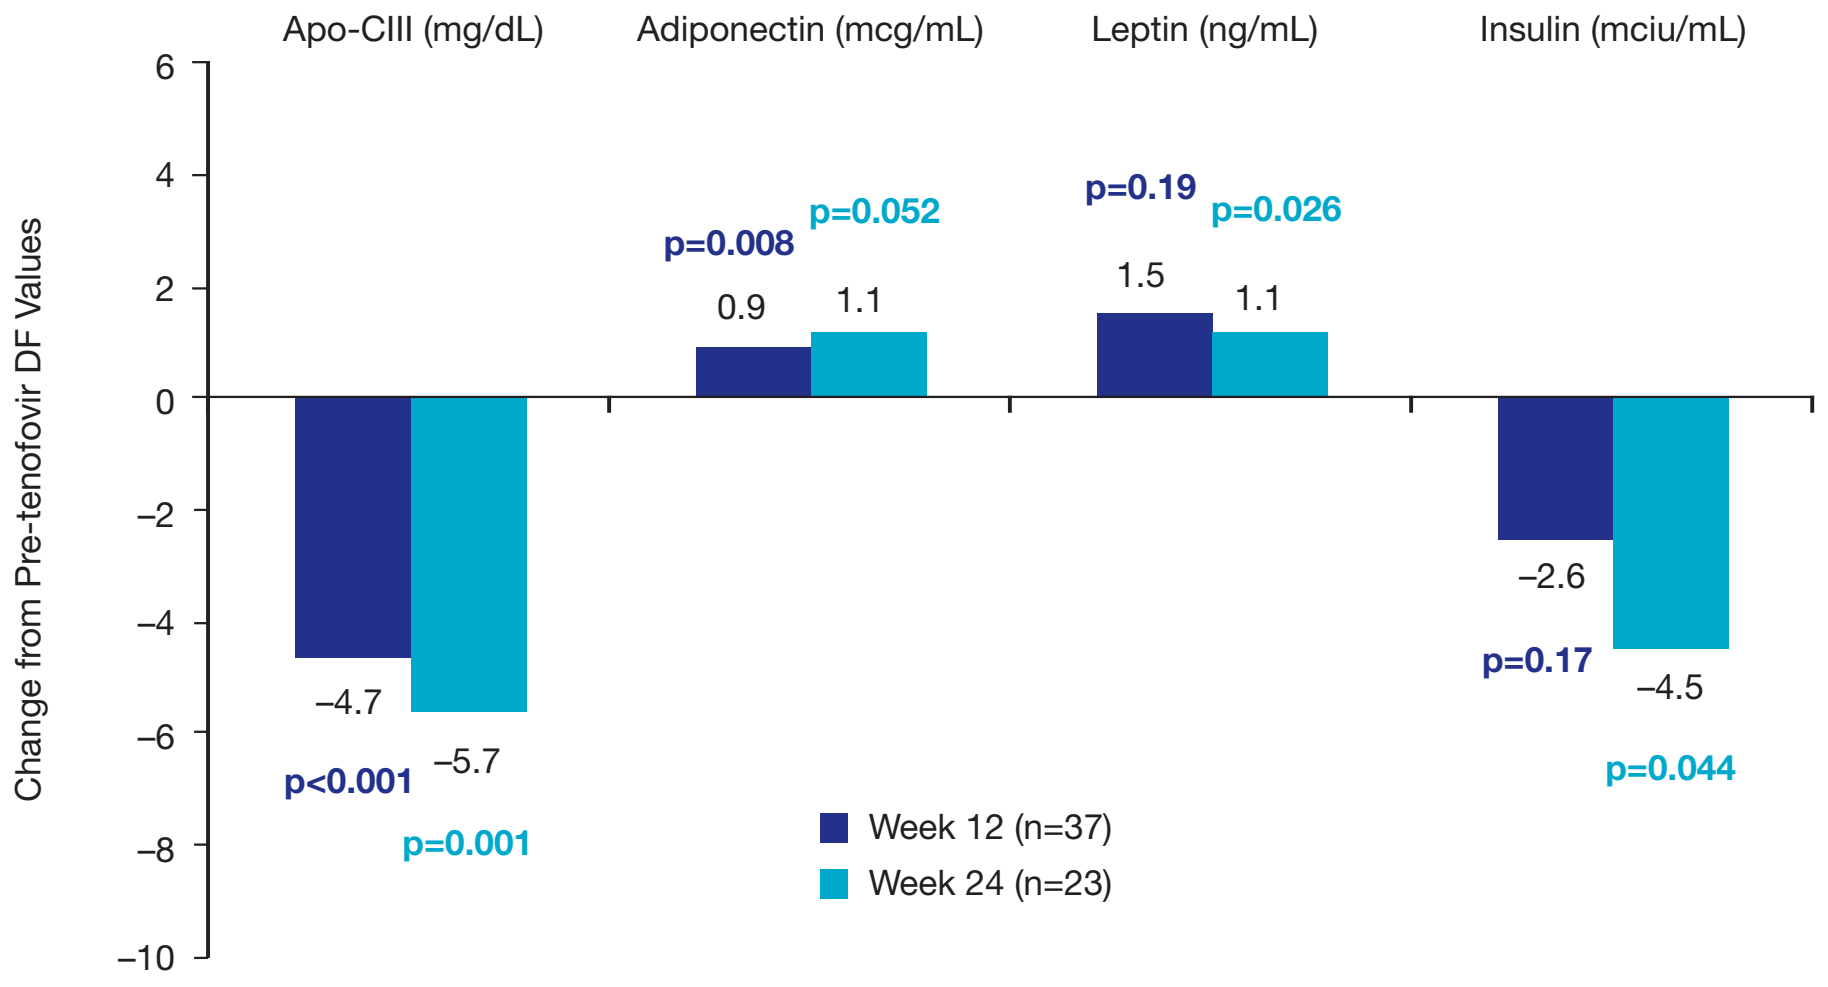
Apo-CIII (mg/dL)
Adiponectin (mcg/mL)
Leptin (ng/mL)
Insulin (mciu/mL)
Change from Pre-tenofovir DF Values
6
4
2
0
–2
–4
–6
–8
–10
–4.7
p<0.001
–5.7
p=0.001
p=0.008
0.9
p=0.052
1.1
p=0.19
1.5
p=0.026
1.1
–2.6
p=0.17
–4.5
p=0.044
Week 12 (n=37)
Week 24 (n=23)

p-values represent comparison with values prior to switch to tenofovir DF

- Mean HOMA-IR prior to the switch to tenofovir DF was 2.8. After 12 weeks, mean HOMA-IR index decreased to 2.2 (p=0.270), with a trend toward continued decreases in those treated for 24 weeks (decrease from 2.6 to 1.7, p=0.071).
- No subject had fasting glucose levels >126 mg/dL (7 mmol/L) either immediately prior to or 12 weeks after the switch to tenofovir DF, and no subject in this analysis was using anti-diabetic medications.
- Measurements of fasting glucose, Apo-AI, hsCRP, and homocysteine did not demonstrate significant changes following the switch to tenofovir DF.
- Mean baseline hsCRP level was 0.24 mg/dL prior to the switch to tenofovir DF and 0.23 mg/dL 12 weeks after the switch (p=0.92). Only 1 subject had hsCRP >1 mg/dL prior to the switch to tenofovir DF and 1 subject had hsCRP >1 mg/dL after 12 weeks of tenofovir DF treatment.

Summary and Discussion

- In this study, subjects who replaced stavudine with tenofovir DF after more than 6 years of antiretroviral therapy with a stable LPV/r-based regimen maintained virologic and immunologic responses.
- These subjects demonstrated significant metabolic improvements following the substitution of stavudine with tenofovir DF, suggesting that the metabolic effects of stavudine are partly reversible despite several years of treatment with stavudine. Changes included:
 - Significant decreases in fasting total cholesterol and triglycerides through 12 and 24 weeks of tenofovir DF treatment.
 - Significant decrease in apolipoprotein CIII levels through 12 and 24 weeks of tenofovir DF treatment.
 - Significant increase in adiponectin levels through 12 weeks of tenofovir DF treatment, coupled with a significant decrease in fasting insulin levels through 24 weeks and a corresponding trend toward decreases in the HOMA-IR assessment through 24 weeks.
 - Significant increase in leptin levels through 24 weeks of tenofovir DF treatment.
- After more than 6 years of a LPV/r-based antiretroviral regimen, no subject had elevated fasting blood glucose levels (>126 mg/dL or 7 mmol/L). However, stavudine was associated with a decrease in insulin sensitivity which was observed only upon stavudine discontinuation when serum insulin decreased, adiponectin levels increased, and HOMA-IR trended downward.
- The small but significant increases in leptin levels are suggestive of possible improvement of stavudine-induced lipoatrophy.

Discussion/Conclusions

- Fasting total cholesterol and triglyceride levels decreased significantly following a switch from stavudine to tenofovir DF in patients who had received LPV/r + stavudine + lamivudine for >6 years, suggesting that the lipid elevations noted in these subjects were related in part to stavudine.
- No subject had elevated fasting blood glucose levels after >6 years of a LPV/r-based regimen.
- Subjects also had low hsCRP levels within the range usually associated with low cardiovascular risk after >6 years of a LPV/r-based regimen.

References

1. Gallant et al. *JAMA*. 2004;292:191-201.
2. Lafeuillade A et al. *JAIDS*. 2003;33:544-546.
3. Domingo P et al. *AIDS*. 2004;18:1475-1478.
4. Garcia-Benayas T et al. *AIDS Res Hum Retroviruses*. 2004;20:1289-1292.
5. Cohn JS et al. *Atherosclerosis*. 2004;177:137-145.
6. Lihn AS et al. *Obes Res*. 2005;6:13-21.
7. Lemieux I et al. *Ann Med*. 2003;35:442-448.
8. Yamauchi T et al. *Nature Medicine*. 1002;7:941-946.
9. Lindegaard B et al. *Clin Exp Immunol*. 2004;135:273-279.
10. Lee GA et al. *JAIDS*. 2004;36:645-647.
11. Nagy GS et al. *CID*. 2003;36:795-802.
12. Zhang F et al. *Vitam Horm*. 2005;71:345-372.
13. Ridker PM. *Am J Cardiol*. 2003;92(suppl):17K-22K.
14. Graham IM et al. *JAMA*. 1997;277:1775-1881.

Acknowledgements

We would like to acknowledge the contributions of the study coordinators and study participants who made this study possible.

05ABTB1211