Poster PE7.3/4

Antiviral Activity of Lopinavir/ritonavir-Based Regimens in Subjects with CD4 Cell Counts Below 25 Cells/mm^3

M King[1], M Johnson[2], M Youle[2], S Brun[1], G Hanna[1]
[1]Abbott, Abbott Park, IL, United States of America; [2]Royal Free Centre for HIV Medicine, London, United Kingdom

Background

Studies of antiretroviral-naïve subjects sometimes exclude those with the lowest CD4 cell counts (1, 2) or do not enroll large numbers of such subjects. Thus, the antiviral activity of HAART in antiretroviral-naïve subjects with very advanced HIV disease is not well characterized. One study has demonstrated higher virologic failure rates in efavirenz- or nevirapine-treated subjects with baseline CD4 cell count <25 cells/mm^3 (3).

Studies of lopinavir/ritonavir (LPV/r)-based regimens have suggested no reduction in efficacy in antiretroviral-naïve subjects with baseline CD4 cell counts <200 cells/mm^3 compared to those with higher baseline CD4 cell counts through 2 or more years of therapy (4, 5). Further, a large comparative study indicated good antiviral activity among subjects with baseline CD4 cell counts <50 cells/mm^3, in contrast to significantly poorer responses among nelfinavir-treated subjects with such low CD4 cell counts (6).

Sample sizes of individual studies of LPV/r-based regimens in antiretroviral-naïve subjects have not been sufficient to examine CD4 cell count cutoffs lower than 50 cells/mm^3. Therefore, we conducted a combined analysis of 4 studies of LPV/r-based regimens to assess response in subjects with baseline CD4 cell count <25 cells/mm^3.

Methods

Data from 654 subjects were combined from 4 randomized clinical trials of LPV/r conducted in antiretroviral-naïve subjects (Table 1).

Table 1. Study Designs

Study	Subjects Receiving LPV/r-Based Regimens	Design	Duration*
M97-720	100	3 blinded doses for 48 weeks, open-label LPV/r 400/100 mg BID thereafter, NRTIs: d4T+3TC BID	7 years (360 weeks)
M98-863	326	blinded, active-controlled (vs. nelfinavir), LPV/r 400/100 mg BID, NRTIs: d4T+3TC BID	60 weeks
M99-056	38	open-label, pilot comparison of QD (800/200 mg) vs. BID (400/100 mg) LPV/r, NRTIs: d4T+3TC BID	96 weeks
M02-418	190	open-label, QD (800/200 mg) vs. BID (400/100 mg) LPV/r, NRTIs: TDF+FTC QD	96 weeks

* Minimum duration of follow-up for all subjects completing the study

The following baseline disease categories were assessed:

- Baseline CD4 cell count: (<25, 25–49, 50–199, 200–349, 350–499, 500 or more cells/mm^3).
- Baseline plasma HIV-1 RNA level (>300,000; 100,000 to 300,000; 30,000 to 100,000; <30,000 copies/mL).

Two complementary analyses of antiviral activity were conducted. In the first, all discontinuations are considered treatment failures. In the second, subjects who discontinue with undetectable HIV-1 RNA levels are censored as of the final visit.

- Proportion of subjects with HIV-1 RNA <50 copies/mL at Week 48 (intent-to-treat, noncompleter=failure method)
 - Subjects with missing values at Week 48 were considered non-responders unless the immediately preceding and immediately following values were <50 copies/mL.
 - Response in each baseline disease category was compared to the reference category (CD4 count >500 cells/mm^3 or HIV-1 RNA level <30,000 copies/mL) using Fisher's exact test.

- The time to loss of virologic response through Week 48. Loss of virologic response was defined to occur on Day 0, if the subject never achieved HIV-1 RNA <400 copies/mL; or on the day of the first of 2 consecutive rebound HIV-1 RNA levels above 400 copies/mL; or on the day of the final visit, if it represented the first rebound value above 400 copies/mL.
 - Subjects who discontinue or complete the study without a loss of virologic response are censored as of their final visit.
 - Subjects in each baseline disease category were compared to subjects in the reference category (CD4 cell count >500 cells/mm³ or HIV-1 RNA level <30,000 copies/mL) using the Cox proportional hazards model.

Results

Baseline Disease Characteristics

- The distribution of baseline HIV-1 RNA levels and CD4 cell counts is shown in Table 2.

Table 2. Baseline Disease Characteristics

CD4 Cell Count (cells/mm³)	
500 or more	94 (14%)
350–499	114 (17%)
200–349	159 (24%)
50–199	163 (25%)
25–49	49 (7.5%)
<25	74 (11%)
missing	1 (<1%)
Mean (SD)	271 (217)
Median (IQR)	241 (89–395)
HIV-1 RNA levels (copies/mL)	
<30,000	196 (30%)
30,000 to 100,000	163 (25%)
100,000 to 300,000	158 (24%)
>300,000	137 (21%)
Mean (SD) (log_{10} copies/mL)	4.9 (0.7)
Median (IQR)	4.9 (4.4–5.4)

Antiviral Activity

- At week 48, no significant differences between baseline CD4 cell count categories (Figure 1) or baseline HIV-1 RNA categories (Figure 2) were observed.

Figure 1. Week 48 HIV-1 RNA <50 copies/mL by Baseline CD4 Cell Count

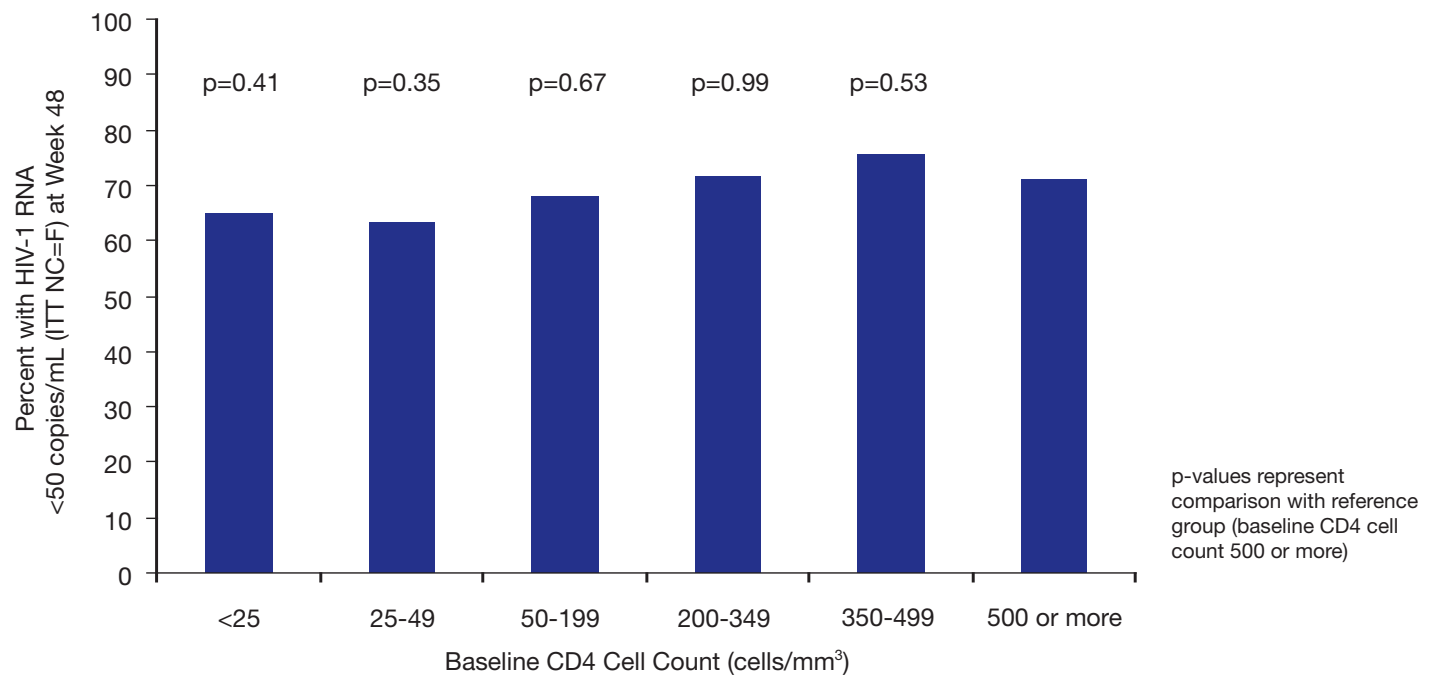
Percent with HIV-1 RNA <50 copies/mL (ITT NC=F) at Week 48
100
90
80
70
60
50
40
30
20
10
0
p=0.41
p=0.35
p=0.67
p=0.99
p=0.53
<25
25-49
50-199
200-349
350-499
500 or more
Baseline CD4 Cell Count (cells/mm³)
p-values represent comparison with reference group (baseline CD4 cell count 500 or more)

Figure 2. Week 48 HIV-1 RNA <50 copies/mL by Baseline HIV-1 RNA Level

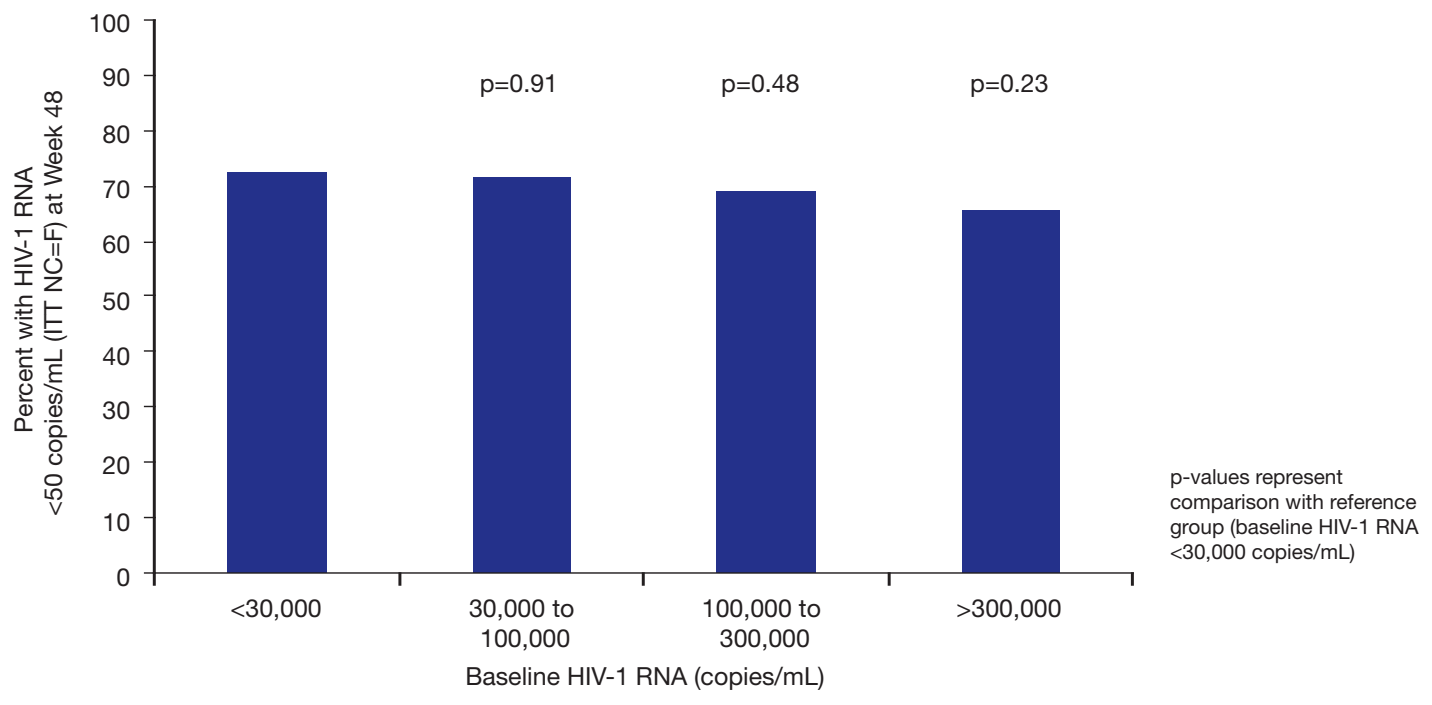
Percent with HIV-1 RNA <50 copies/mL (ITT NC=F) at Week 48
100
90
80
70
60
50
40
30
20
10
0
p=0.91
p=0.48
p=0.23
<30,000
30,000 to 100,000
100,000 to 300,000
>300,000
Baseline HIV-1 RNA (copies/mL)
p-values represent comparison with reference group (baseline HIV-1 RNA <30,000 copies/mL)

- Through 48 weeks, no significant differences in the time to loss of virologic response were observed among baseline CD4 cell count or HIV-1 RNA level categories (Table 3).
- Kaplan-Meier estimates of the time to loss of virologic response are displayed by CD4 cell count in Figure 3 and by HIV-1 RNA level in Figure 4.

Table 3. Time to Loss of Virologic Response Through Week 48

Category	Hazard Ratio (95% CI)	p-value
CD4 cell count (cells/mm³)		
500 or more	reference	n/a
350–499	0.7 (0.3, 1.4)	0.27
200–349	0.6 (0.3, 1.2)	0.15
50–199	0.7 (0.3, 1.3)	0.22
25–49	0.8 (0.3, 1.9)	0.57
<25	1.0 (0.5, 2.1)	0.95
HIV-1 RNA levels (copies/mL)		
<30,000	reference	n/a
30,000 to 100,000	0.9 (0.5, 1.6)	0.72
100,000 to 300,000	1.3 (0.7, 2.2)	0.42
>300,000	0.8 (0.4, 1.6)	0.57

Figure 3. Time to Loss of Virologic Response by Baseline CD4 Cell Count

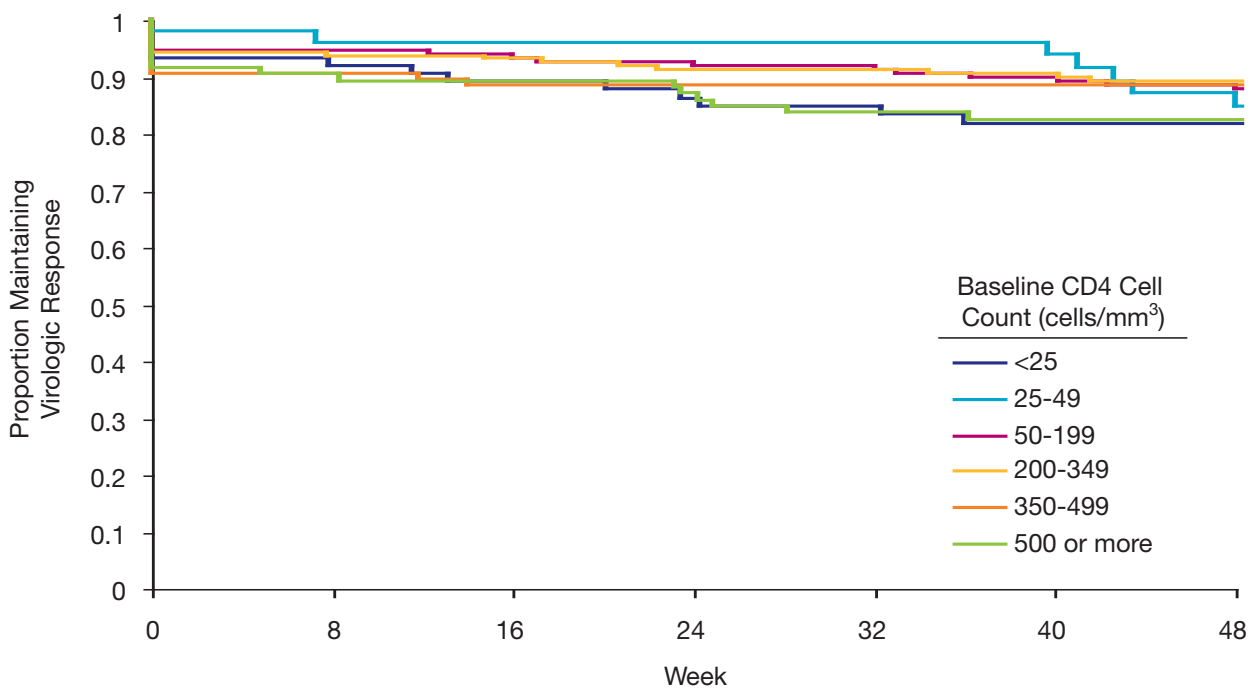
Proportion Maintaining Virologic Response
1
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0
0
8
16
24
32
40
48
Week
Baseline CD4 Cell Count (cells/mm3)
<25
25-49
50-199
200-349
350-499
500 or more

Figure 4. Time to Loss of Virologic Response by Baseline HIV-1 RNA Level

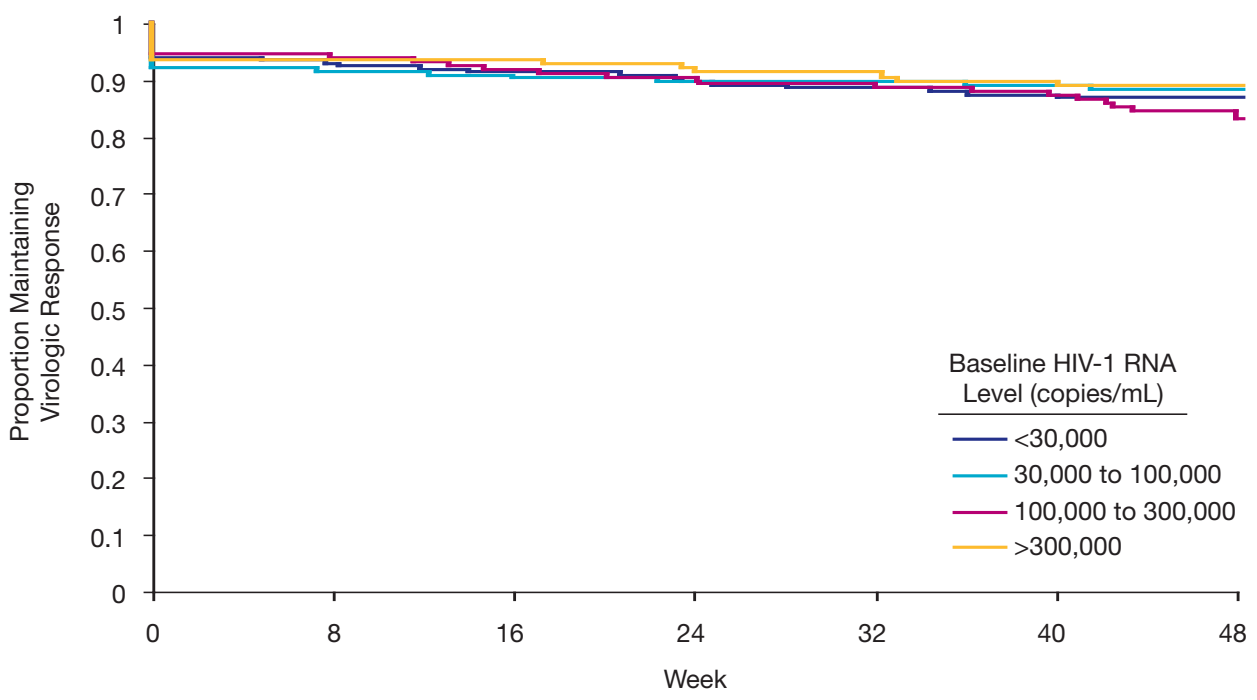
Proportion Maintaining Virologic Response
1
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0
0
8
16
24
32
40
48
Week
Baseline HIV-1 RNA Level (copies/mL)
<30,000
30,000 to 100,000
100,000 to 300,000
>300,000

Immunologic Response

- The change from baseline to Week 48 in CD4 cell count was generally comparable across baseline CD4 cell count categories (Figure 5). With the exception of the group with baseline CD4 cell count 50–199 cells/mm^3, mean increases in CD4 cell counts were not significantly different from those in the reference group.
- At Week 48, 44% of those with baseline CD4 cell count <25 cells/mm^3 had CD4 cell count >200 cells/mm^3, as did 62% of those with baseline CD4 cell count between 25–49 cells/mm^3. No subject had CD4 cell count <50 cells/mm^3 at Week 48.

Figure 5. Week 48 Mean Change from Baseline in CD4 Cell Count by Baseline CD4 Cell Count

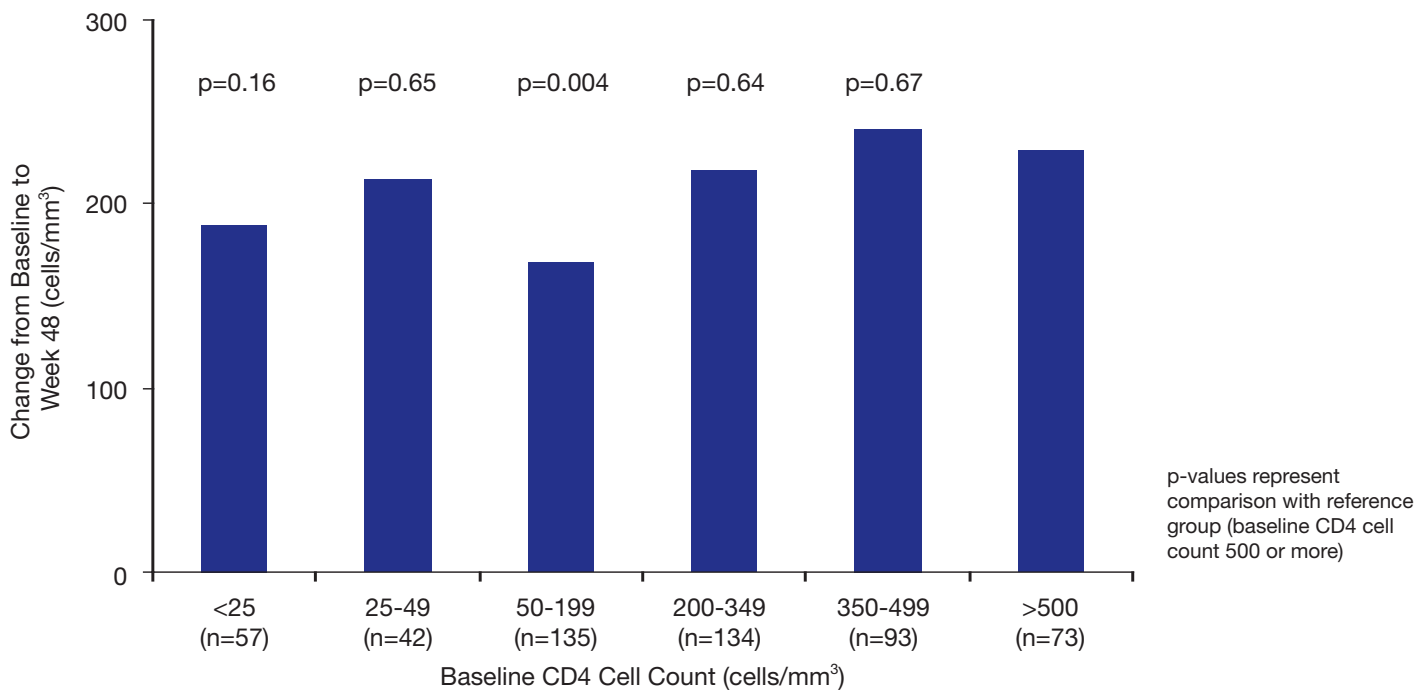
300
200
100
0
Change from Baseline to Week 48 (cells/mm3)
p=0.16
p=0.65
p=0.004
p=0.64
p=0.67
<25 (n=57)
25-49 (n=42)
50-199 (n=135)
200-349 (n=134)
350-499 (n=93)
>500 (n=73)
Baseline CD4 Cell Count (cells/mm3)
p-values represent comparison with reference group (baseline CD4 cell count 500 or more)

Sensitivity Analyses

- To ensure that the finding of no statistically significant differences was not due to the choice of reference group, we assessed the time to loss of virologic response by all possible pairwise comparisons of the baseline CD4 cell count and HIV-1 RNA level categories
 - CD4 cell count: Each of the 6 categories was compared to each other category, resulting in 15 possible pairwise comparisons. None of the 15 comparisons demonstrated a statistically significant difference between categories (p>0.14 for all comparisons)
 - HIV-1 RNA level: Each of the 4 categories was compared to each other category, resulting in 6 possible pairwise comparisons. None of the 6 comparisons demonstrated a statistically significant difference between categories (p>0.18 for all comparisons).
- To ensure that observed results were not a manifestation of insufficient sample size, we combined adjacent categories and assessed the time to loss of virologic response. Specifically, subjects were dichotomized and compared by various CD4 cell counts (<25 vs. 25 or more cells/mm^3, <50 vs. 50 or more cells/mm^3, <200 vs. 200 or more cells/mm^3, etc.) and baseline HIV-1 RNA levels (<30,000 vs. >30,000 copies/mL, <100,000 vs. >100,000 copies/mL, <300,000 vs. >300,000 copies/mL). No statistically significant differences were observed for any of these comparisons (p>0.20 for each).

Discussion/Conclusions

- Studies of antiretroviral-naïve subjects often exclude subjects with very low CD4 counts. In one study with no lower bound on entry CD4 cell counts, NNRTI-based regimens demonstrated a higher risk of virologic failure among subjects with baseline CD4 cell count <25 cells/mm^3.
- In contrast, in the current analysis of LPV/r-treated subjects, no significant reduction in efficacy was observed among subjects with very low CD4 cell counts.
- Similarly, antiviral activity of LPV/r-based regimens was not associated with baseline HIV-1 RNA level.
- These findings may have implications for the choice of initial treatment of patients who present with advanced HIV disease.

References

1. S Staszewski, et al. *N Engl J Med* 1999; 341:1865–73.
2. K Squires, et al. *J Acquir Immune Defic Syndr* 2004;36:1011–9.
3. F van Leth, et al. 11th CROI (2004), Abstract 550.
4. M Thompson, et al. 8th ECCATh (2001), Abstract 105.
5. V Yeh, et al. 44th ICAAC (2004), Abstract H-570.
6. M King, et al. *J Infect Dis* 2004;190:280–4.

05ABTB1140 #2