Poster H-835

43rd Interscience Conference on Antimicrobial Agents and Chemotherapy (ICAAC), Chicago, IL, USA
September 14–17, 2003

DURABILITY OF RESPONSE TO ENFUVIRTIDE THROUGH 48 WEEKS IN THE TORO TRIALS

Trottier B[1], Arastéh K[2], Henry K[3], Katlama C[4], Lazzarin A[5], Montaner J[6], DeMasi R[7], Delehanty J[7], Chung J[8] and Salgo M[8] for the TORO Study Team

[1]Clinique Médicale l'Actuel, Montréal, PQ, Canada; [2]EPIMED, Berlin, Germany; [3]HCMC, Minneapolis, MN, USA; [4]Pitié-Salpêtrière Hospital, Paris, France; [5]IRCCS S. Raffaele, Milan, Italy; [6]UBC, Vancouver, BC, Canada; [7]Trimeris, Durham, NC, USA; [8]Roche, Nutley, NJ, USA

Benoit Trottier, MD
Clinique médicale l'Actuel
1001 de Maisonneuve, suite 1130
Montréal, Qc, H2L 4P9
Canada

Ph: +1 514-524-3642
Fax: +1 514-524-3831
E-mail: trottier@lactuel.ca

Background

- Enfuvirtide (ENF) is the first in a new class of antiretroviral (ARV) agents, the HIV-1 fusion inhibitors, which targets HIV gp41 and blocks the virus from entering host cells. The safety and efficacy of chronically administered ENF has been demonstrated in Phase II and III clinical studies.[1–6]
- In the Phase III TORO 1 and TORO 2 trials, patients who had previous treatment with and/or documented resistance to one or more agents from each of the other three classes of ARVs were randomized to receive either enfuvirtide (ENF; 90 mg sc *BID*) plus an optimized background (OB) regimen (based on ARV treatment history and baseline resistance testing) or OB alone. All patients had a plasma HIV-1 RNA level of ≥ 5000 copies/mL.
- Primary analyses of both TORO 1 and TORO 2 at 24 weeks demonstrated that the addition of ENF to OB provided significantly greater viral suppression and immunologic benefit compared to OB alone. A pre-planned analysis of the pooled 24 week data indicated that patients on the ENF + OB arm were twice as likely to achieve a reduction in HIV-1 RNA to below detection ($P < 0.0001$ for both < 400 copies/mL and < 50 copies/mL) and achieve double the increase in CD4+ cell count ($P < 0.0001$).[6]
- Initial analyses of results from the week 48 combined TORO database described the superior efficacy of ENF + OB compared to OB alone.[7]
- While the week 48 analyses support long-term benefit, we report here additional evaluations that provide more details about the durability of response associated with ENF.
- Safety analyses at week 48 are reported elsewhere.[8]

Methods

Analysis of virologic response

- Durability of response was evaluated by methods that included:
 - i) Assessment of overall response (treatment benefit) [e.g. viral load < 400 copies/mL] at week 48
 - ii) Patients maintaining a response from week 24 to 48
 - iii) Number of new responders at week 48 (after week 24)
 - iv) Time to protocol-defined virologic failure.
- Virologic failure, based on two consecutive values, was prospectively defined as an inadequate virologic response or loss of virologic response, according to one of three criteria:
 - i) < 0.5 $\log_{10}$ copies/mL reduction in baseline viral load starting at weeks 6 and 8
 - ii) < 1.0 $\log_{10}$ copies/mL reduction in baseline viral load starting at weeks 14 and 16
 - iii) > 1.0 $\log_{10}$ copies/mL increase in HIV-1 RNA after a nadir of ≥ 2 $\log_{10}$ copies/mL (average of the two lowest viral load values) below baseline.
- Patients initially randomized to OB alone who experienced virologic failure after week 8 were allowed to switch to receive ENF plus a revised OB regimen.

Analysis methods

- As pre-planned, since the results were consistent across TORO 1 and TORO 2, data from the two studies were pooled. In this poster pooled results are presented.
- Responders at week 48 included week 24 responders maintaining their response and patients meeting response criteria after week 24 (new responders).
- The time to virologic failure was estimated using the method of Kaplan–Meier plot and log-rank test.
- For change in HIV-1 RNA and CD4+ cell count, discontinuations and virologic failure carried the average of the last two observations forward (LOCF). Additional sensitivity analyses were conducted considering discontinuations and switch as failures (no change from baseline).
- For responder analysis (by < 50 copies/mL, < 400 copies/mL and ≥ 1.0 $\log_{10}$ change from baseline), discontinuations and virologic failure were considered as failures.
- For time to virologic failure, non-virologic failure and discontinuations were censored.

Results

Virologic and immunologic response

- Baseline characteristics of the 995 evaluable patients are shown in **Table 1**.

Table 1. Combined TORO 1 and TORO 2: pooled baseline characteristics and prior ARV experience of study populations (ITT)

	ENF + OB (n = 661)	OB (n = 334)
Baseline HIV-1 RNA (median, $\log_{10}$ copies/mL)	5.2	5.1
Baseline CD4+ cell count (median, cells/mm^3)	88	97
Number of prior ARVs (median)	12	12
Years since initiating ARVs (median)	7	7
Prior NRTI (median duration, years)	6.3	6.3
Prior NNRTI (median duration, years)	1.4	1.5
Prior PI (median duration, years)	3.8	4.0

- At week 48, a significantly greater proportion of patients randomized to ENF + OB reached each responder category (a ≥ 1 $\log_{10}$ drop from baseline, < 400 and < 50 copies/mL) compared to OB alone (**Figure 1**; $P < 0.0001$ for all comparisons).

Figure 1. Combined TORO 1 and TORO 2: categoric responder analysis at weeks 24 and 48 (ITT, discontinuations + virologic failure = failure)*

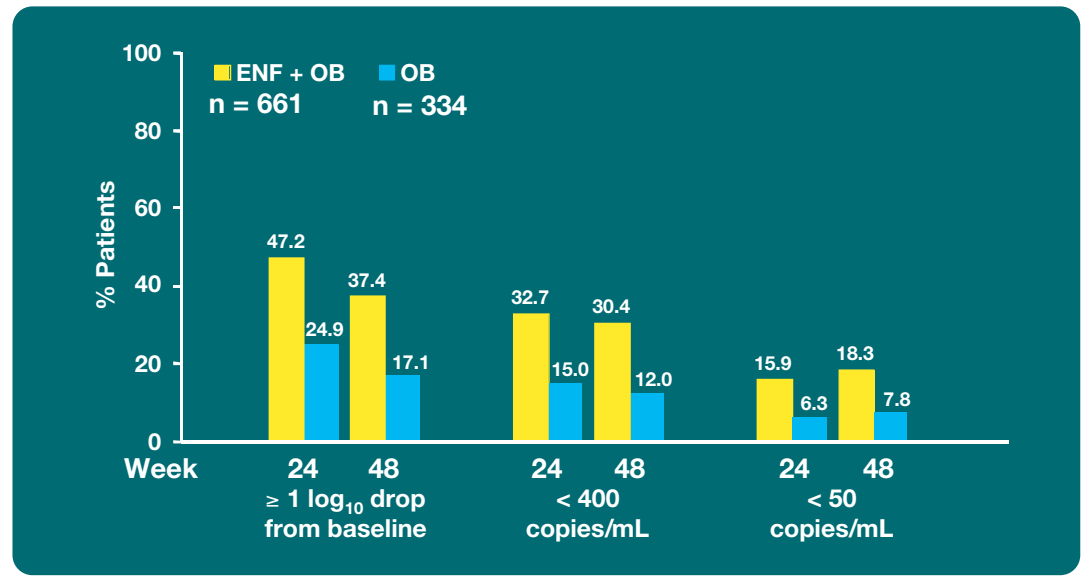
ENF + OB n = 661
OB n = 334
% Patients
47.2 24.9 37.4 17.1
32.7 15.0 30.4 12.0
15.9 6.3 18.3 7.8
Week 24 48 24 48 24 48
≥ 1 log10 drop from baseline
< 400 copies/mL
< 50 copies/mL

* All ENF + OB *vs.* OB comparisons were statistically significant ($P < 0.0001$)

- At week 48 (ITT, LOCF), patients randomized to ENF + OB had experienced a mean reduction from baseline viral load of 1.48 $\log_{10}$ copies/mL, compared to 0.63 $\log_{10}$ copies/mL for patients randomized to OB alone **(Figure 2a)**.
- At week 48, patients randomized to ENF + OB had experienced a mean increase in CD4+ count of 91 cells/mm^3 compared to 45 cells/mm^3 in the OB alone arm **(Figure 2b)**.
- The sensitivity analyses for change from baseline in viral load and CD4+ cell count considering discontinuations and switch as failures (no change from baseline) showed slightly greater treatment differences that were also statistically significant (change in HIV-1 RNA –0.895 $\log_{10}$ copies/mL, $P < 0.0001$; change in CD4+ count 66.6 cells/mm^3, $P < 0.0001$; both favoring ENF + OB).

Figure 2. Combined TORO 1 and TORO 2: efficacy at 24 and 48 weeks
Figure 2a. Plasma HIV-1 RNA adjusted means change from baseline (ITT, LOCF)

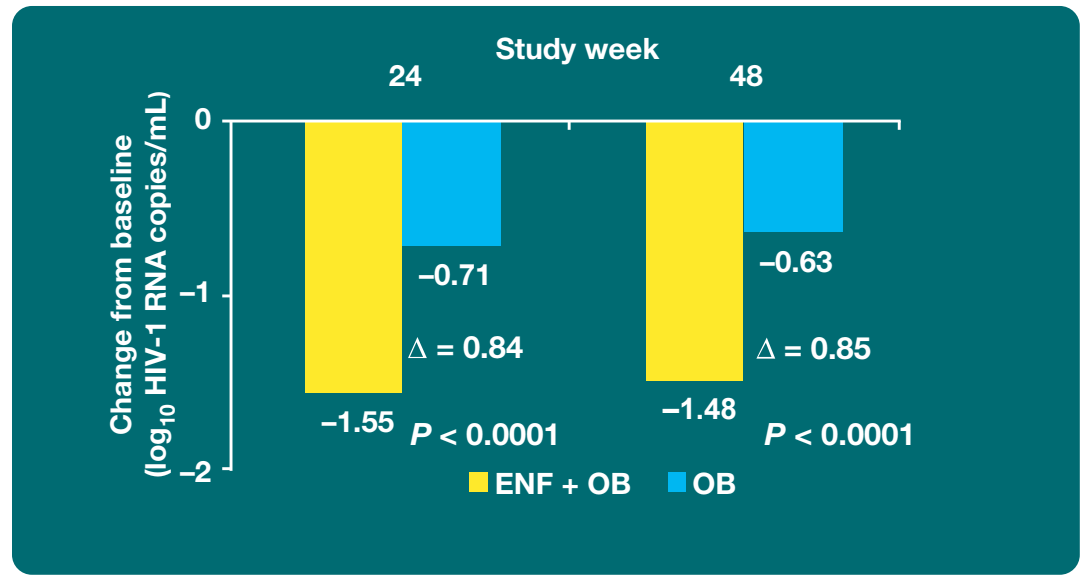
Study week
24
48
Change from baseline (log10 HIV-1 RNA copies/mL)
–1.55
–0.71
Δ = 0.84
P < 0.0001
–1.48
–0.63
Δ = 0.85
P < 0.0001
ENF + OB
OB

Figure 2b. CD4+ cell count adjusted means change from baseline (ITT, LOCF)

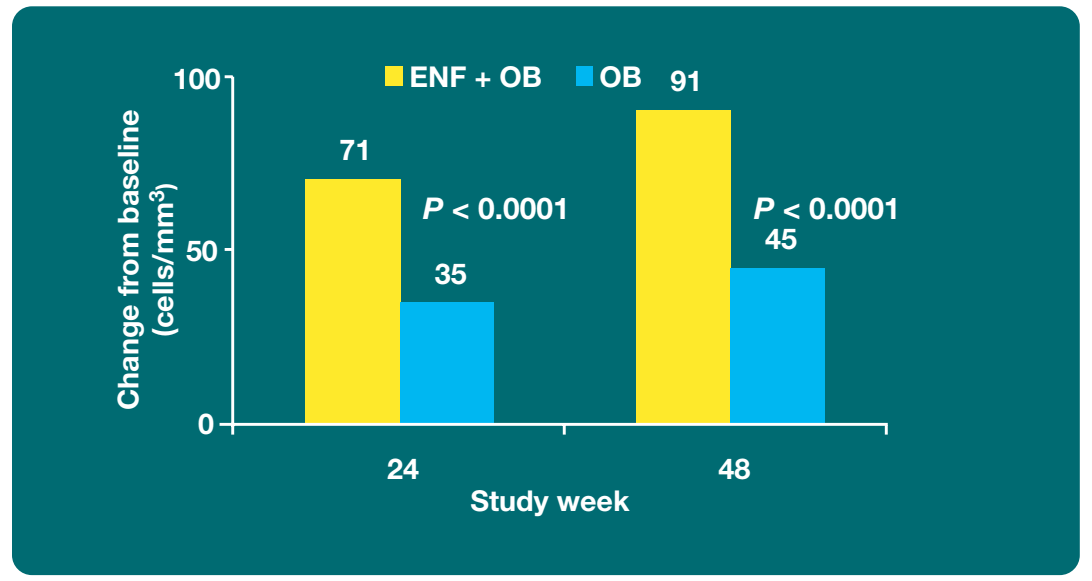
ENF + OB
OB
Change from baseline (cells/mm3)
71
35
P < 0.0001
91
45
P < 0.0001
24
48
Study week

- Across both treatment groups, the proportion of week 24 responders who maintained their response at week 48 was high **(Figure 3)**.

Figure 3. Combined TORO 1 and TORO 2: proportion of week 24 responders who maintained their response at week 48

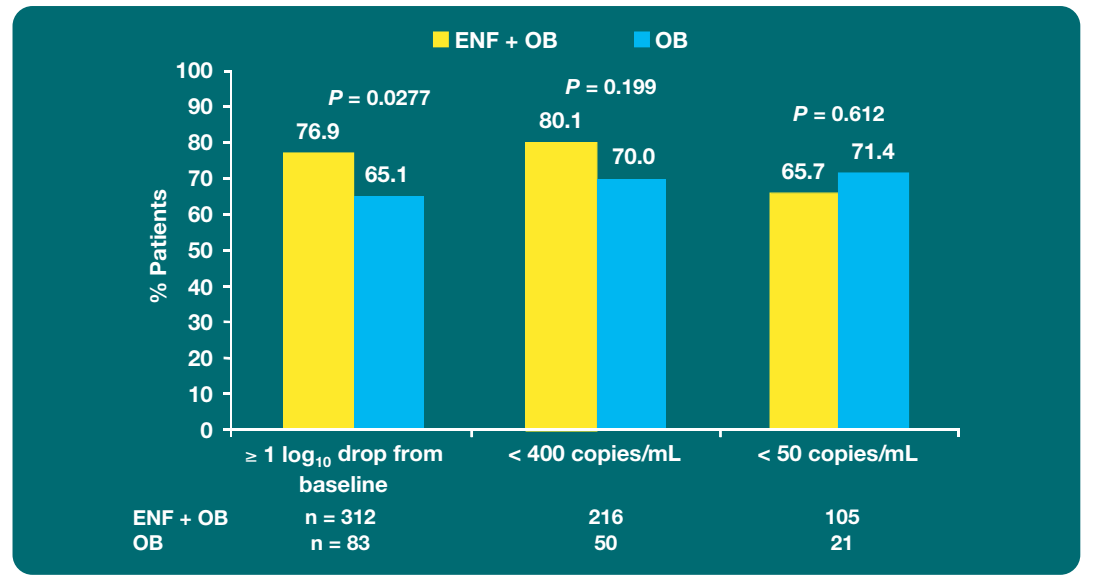
ENF + OB
OB
% Patients
P = 0.0277
76.9
65.1
P = 0.199
80.1
70.0
P = 0.612
65.7
71.4
≥ 1 log10 drop from baseline
< 400 copies/mL
< 50 copies/mL
ENF + OB n = 312 216 105
OB n = 83 50 21

- For response categories < 50 copies/mL and < 400 copies/mL, there were a greater number of new responders at week 48 in the ENF + OB arm compared to OB alone, whereas for the ≥ 1.0 $\log_{10}$ drop from baseline category the numbers of new responders were similar **(Table 2)**.

Table 2. Combined TORO 1 and TORO 2: week 24 responders maintaining response and new responders at week 48

HIV-1 RNA response category at week 48	**ENF + OB n = 661**	**OB n = 334**
< 50 copies/mL	**121 (18.3%)**	**26 (7.8%)**
Maintained week 24 response	69 (10.4%)	15 (4.5%)
New response	52 (7.9%)	11(3.3%)
< 400 copies/mL	**201 (30.4%)**	**40 (12.0%)**
Maintained week 24 response	173 (26.2%)	35 (10.5%)
New response	28 (4.2%)	6 (1.8%)
≥ 1.0 $\log_{10}$ drop from baseline	**247 (37.4%)**	**57 (17.1%)**
Maintained week 24 response	240 (36.3%)	54 (16.2%)
New response	7 (1.1%)	3 (0.9%)

- The median time to virologic failure was three times longer on ENF + OB (32 weeks) compared to OB alone (11 weeks) ($P < 0.0001$) **(Figure 4)**.

Figure 4. Combined TORO 1 and TORO 2: analysis of time to protocol-defined virologic failure up to week 48 (ITT)

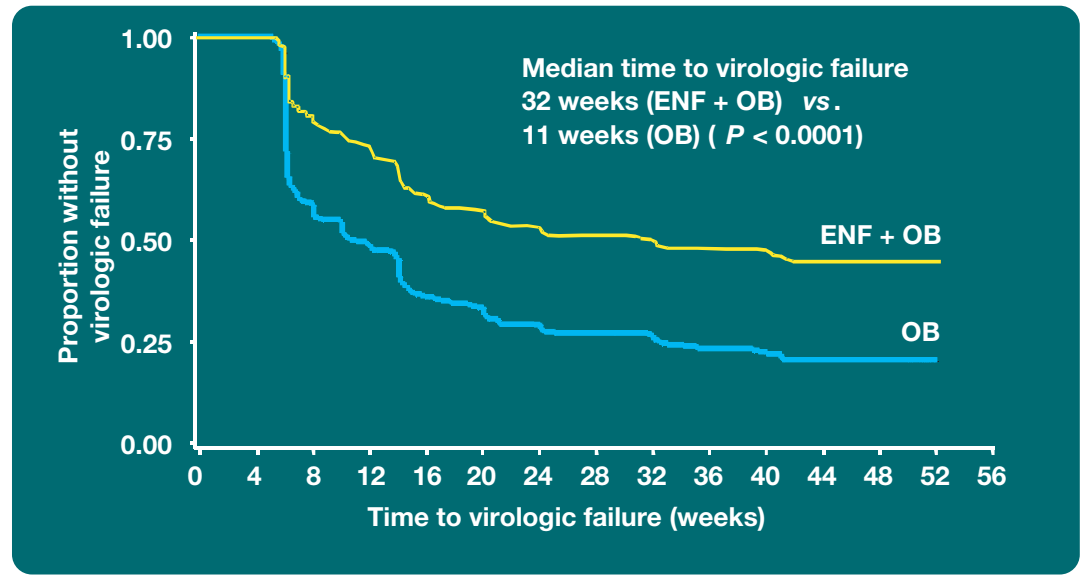
Median time to virologic failure
32 weeks (ENF + OB) vs.
11 weeks (OB) (P < 0.0001)
Proportion without virologic failure
ENF + OB
OB
Time to virologic failure (weeks)

- In both arms, time to virologic failure was longer in patients who experienced fewer prior ARVs **(Figure 5)**.

Figure 5. Combined TORO 1 and TORO 2: Kaplan–Meier plots of time to virologic failure by number of prior ARVs

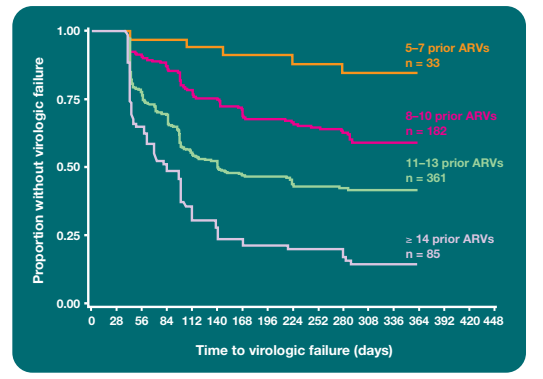
Proportion without virologic failure
5–7 prior ARVs n = 33
8–10 prior ARVs n = 182
11–13 prior ARVs n = 361
≥ 14 prior ARVs n = 85
Time to virologic failure (days)

Figure 5a. Analysis of time to virologic failure up to week 48 for ENF + OB patients by number of prior ARVs (ITT)

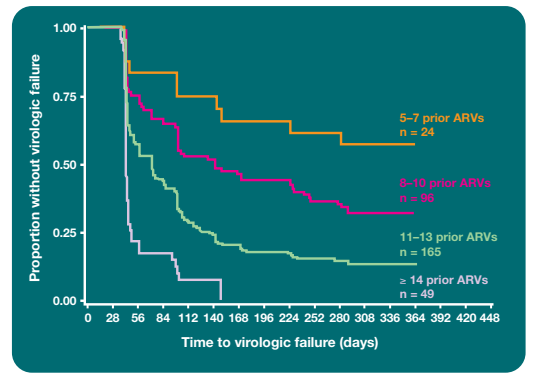
Proportion without virologic failure
5–7 prior ARVs n = 24
8–10 prior ARVs n = 80
11–13 prior ARVs n = 165
≥ 14 prior ARVs n = 49
Time to virologic failure (days)

Figure 5b. Analysis of time to virologic failure up to week 48 for OB patients by number of prior ARVs (ITT)

Median time to virologic failure (days)

ENF + OB	OB
5–7 prior ARVs – cannot be estimated	**5–7 prior ARVs – cannot be estimated**
8–10 prior ARVs – cannot be estimated	**8–10 prior ARVs – 141**
11–13 prior ARVs – 141	**11–13 prior ARVs – 71**
≥ 14 prior ARVs – 85	**≥ 14 prior ARVs – 43**

- **Figure 6** shows:
 - Subgroup analyses by the number of active ARVs in the OB regimen (genotypic sensitivity score; GSS) demonstrated virologic benefit of ENF + OB over OB alone across the spectrum of response, including those with zero or one active agents in the OB regimen (GSS = 0 or 1)
 - Virologic response increased with increasing number of active agents in the OB regimen (increasing GSS)
 - ENF + OB with one active agent provided benefit (28.9% < 400 copies/mL) similar to or greater than OB alone with two, three, four or five active ARVs in the OB regimen (15.1%, 18.0%, 28.6% or 12.5% below < 400 copies/mL, respectively).

Figure 6. Combined TORO 1 and TORO 2: treatment benefit across individual GSS subgroups for responders with HIV-1 RNA < 400 copies/mL at week 48 (ITT, discontinuations + virologic failure = failure)

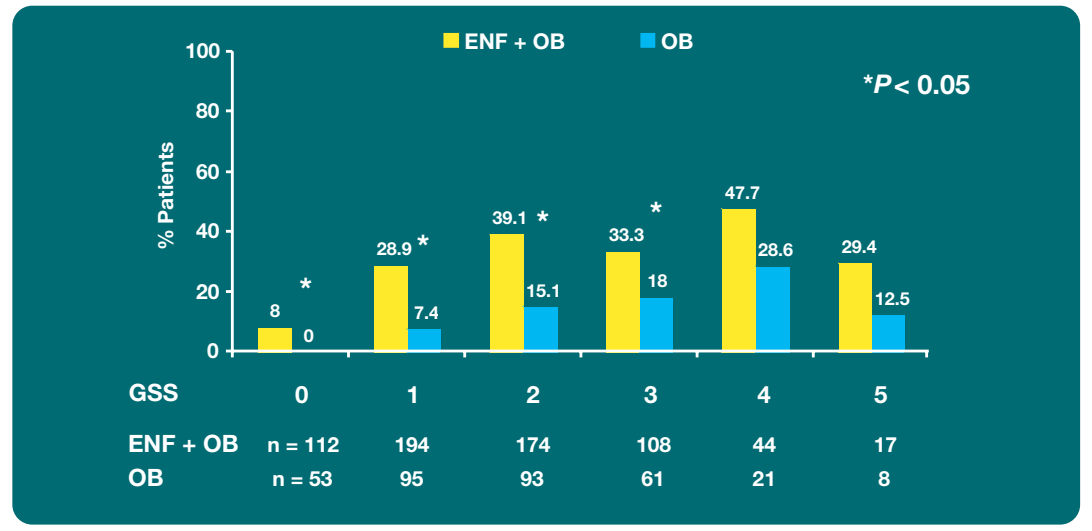
ENF + OB
OB
*P< 0.05
% Patients
8 0
28.9 7.4
39.1 15.1
33.3 18
47.7 28.6
29.4 12.5
GSS 0 1 2 3 4 5
ENF + OB n = 112 194 174 108 44 17
OB n = 53 95 93 61 21 8

- Patients receiving ENF + OB had significantly greater increases in CD4+ cell count compared to OB alone for all groups by the number of active ARVs in the OB regimen, including those with zero or one active agents in the OB regimen **(Figure 7)**.

Figure 7. Combined TORO 1 and TORO 2: CD4+ cell count adjusted means change from baseline at week 48 (ITT, LOCF) by baseline GSS

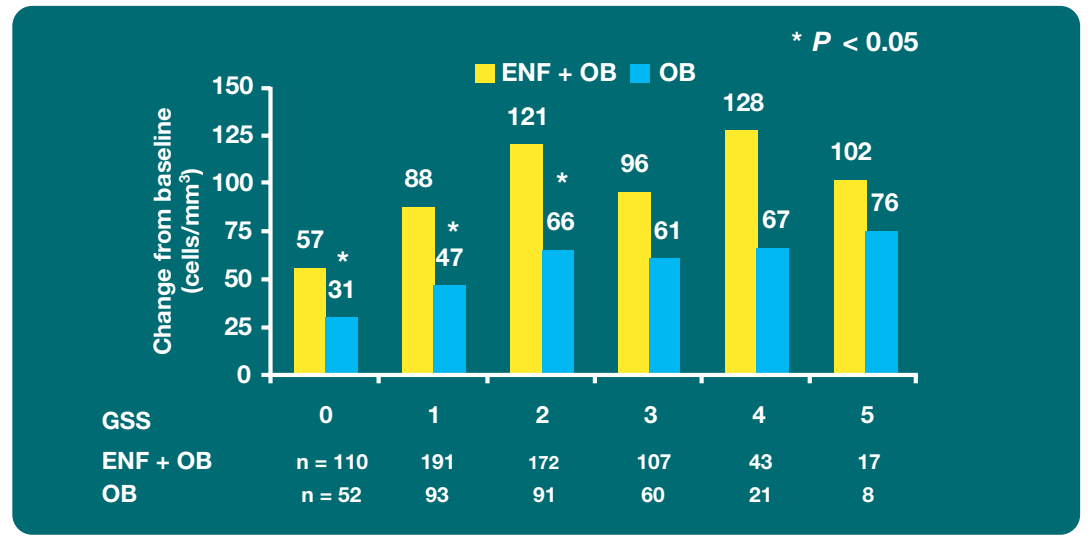
* P < 0.05
ENF + OB
OB
Change from baseline (cells/mm3)
57 31
88 47
121 66
96 61
128 67
102 76
GSS 0 1 2 3 4 5
ENF + OB n = 110 191 172 107 43 17
OB n = 52 93 91 60 21 8

Conclusions

- The overall response rate at week 48 demonstrated significantly greater treatment benefit for patients in the ENF + OB arm compared to the OB alone arm, confirming week 24 data.
- Therapy with ENF + OB provided consistent and durable benefit — virologic responses seen at 24 weeks were maintained at 48 weeks by the majority of patients receiving ENF + OB.
- At week 48 the proportion of new responders was higher on ENF + OB compared to OB alone.
- The time to protocol-defined virologic failure was approximately three times longer on ENF + OB compared to OB (32 *vs.* 11 weeks).
- The time to protocol-defined virologic failure across subgroups of patients defined by the number of prior ARVs was longer on ENF + OB compared to OB for all subgroups. In addition, those with fewer prior ARVs had longer time to protocol-defined virologic failure compared to more heavily pretreated patients.
- Subgroup analyses by the number of active ARVs in the OB regimen demonstrated virologic and immunologic benefit of ENF + OB over OB across all subgroups, including those with zero or one active agents.
- The magnitude of response increased with increasing number of active agents in the OB regimen; the proportion with viral load < 400 copies/mL on ENF + OB with one active agent in the OB regimen was similar to or greater than that seen on OB alone with two, three or more active agents.

References

1. Lalezari JP, Eron JJ, Carlson M, *et al*. A Phase II clinical study of the long-term safety and antiviral activity of enfuvirtide-based antiretroviral therapy. *AIDS* 2003; **17**:691–698.
2. Lalezari JP, Henry K, O'Hearn M, *et al.* Enfuvirtide, an HIV-1 fusion inhibitor, for drug-resistant HIV infection in North and South America. *N Engl J Med* 2003; **348**:2175–2185.
3. Lazzarin A, Clotet B, Cooper D, *et al.* Efficacy of enfuvirtide in patients infected with drug-resistant HIV-1 in Europe and Australia. *N Engl J Med* 2003; **348**:2186–2195.
4. Lalezari J, Henry K, O'Hearn M, *et al.* Enfuvirtide (T-20) in combination with an optimized background (OB) regimen vs. OB alone: week 24 response among categories of treatment experience and baseline (BL) HIV antiretroviral (ARV) resistance. *42nd Interscience Conference on Antimicrobial Agents and Chemotherapy*. San Diego, CA, USA, September 27–30 2002; Abstract 3232.
5. Lange J, Lazzarin A, Clotet B, *et al.* Enfuvirtide (T-20) in combination with an optimized background (OB) regimen vs. OB alone: week 24 response among categories of baseline (BL) demographics, treatment experience, and HIV antiretroviral (ARV) resistance. *6th International Congress on Drug Therapy in HIV Infection*. Glasgow, UK, November 17–21 2002; Abstract PL14.3.
6. Delfraissy J-F, Montaner J, Eron J, *et al.* Summary of pooled efficacy and safety analyses of enfuvirtide (ENF) treatment for 24 weeks in TORO 1 and TORO 2 Phase III trials in highly antiretroviral (ARV) treatment experienced patients. *10th Conference on Retroviruses and Opportunistic Infections. Boston, MA, USA,* February 10–14 2003; Abstract 568.
7. Katlama C, Arastéh K, Clotet B, *et al.* Enfuvirtide TORO studies: 48 week results confirm 24 week findings. *2nd IAS Conference on HIV Pathogenesis and Treatment.* Paris, France July 13–16 2003; Abstract LB2.
8. Eron J, Delfraissy J, Kuritzkes D, *et al.* Safety of enfuvirtide (ENF) through 48 weeks of therapy in the TORO trials. *43rd Interscience Conference on Antimicrobial Agents and Chemotherapy*. Chicago, IL, USA, September 14–17 2003; Abstract H-836.

These studies were supported by Roche and Trimeris

Acknowledgements

The Authors would like to express their gratitude to all of the patients that participated in the TORO 1 and 2 trials as well as the TORO Study investigators and the numerous Roche and Trimeris study personnel who have worked on these two studies.

Poster H-835 – 43rd Interscience Conference on Antimicrobial Agents and Chemotherapy (ICAAC), Chicago, IL, USA, September 14–17, 2003

Poster H-835 – 43rd Interscience Conference on Antimicrobial Agents and Chemotherapy (ICAAC), Chicago, IL, USA, September 14–17, 2003

43rd Interscience Conference on Antimicrobial Agents and Chemotherapy (ICAAC), Chicago, IL, USA
September 14–17, 2003

DURABILITY OF RESPONSE TO ENFUVIRTIDE THROUGH 48 WEEKS IN THE TORO TRIALS

Trottier B[1], Arastéh K[2], Henry K[3], Katlama C[4], Lazzarin A[5], Montaner J[6], DeMasi R[7], Delehanty J[7], Chung J[8] and Salgo M[8] for the TORO Study Team

[1]Clinique Médicale l'Actuel, Montréal, PQ, Canada; [2]EPIMED, Berlin, Germany; [3]HCMC, Minneapolis, MN, USA; [4]Pitié-Salpêtrière Hospital, Paris, France; [5]IRCCS S. Raffaele, Milan, Italy; [6]UBC, Vancouver, BC, Canada; [7]Trimeris, Durham, NC, USA; [8]Roche, Nutley, NJ, USA

RO4149/09.03